poster 155 x 110 7.0:Mise en page 1 31/10/11 17:56 Page 1

LB 9

Four weeks of IL-7 (CYT107) added to peginterferon (pegIFN) and ribavirin (RBV) is safe, induces a broad immune response and HCV viral clearance in genotype 1 and 4 patients non responders to pegIFN and RBV (Study ECLIPSE 2)

Habersetzer F[1]., Andreone P[2]., Payen JL[3]., Rouzier R[4]., Attali P[5].,Alric L[6]., Grando V[7]., Tambussi G[8], Serfaty L[9]., Thabut D.[10]., Hézode C[11]., Beq S[12] ., Demol P[12]., Croughs T[12]., Morre M[12]., Marcellin P[13].

[1]Nouvel Hôpital Civil, Strasbourg, France ; [2]Università di Bologna, Bologna, Italy; [3]CHU Montauban, Montauban, France ; [4]Centre CAP Phase I/II, Montpellier, France; [5]Hôpital Kremlin Bicêtre, Kremlin Bicêtre, France; [6]CHU Purpan, Toulouse, France; [7]Hôpital Jean Verdier, Bondy, France; [8]San Raffaele Scientific Institute, Milano, Italy; [9]Hôpital St Antoine, Paris, France; [10]Hôpital Pitié Salpêtrière, Paris, France; [11]Hôpital Henri Mondor, Créteil, France; [12]Cytheris, Issy-les-Moulineaux, France; [13]Hôpital Beaujon, Clichy, France

Backgroud and rational

IL-7 properties match especially well with T cell abnormalities reported in HCV infection:

- **lymphopoietic effect** (thymopoiesis); IL-7 may potently expand the frequency of rare HCV specific CD4 or CD8 T cell precursors
- may **enhance T cells response;** the magnitude of CD4 and CD8 T cell responses, usually blunted in HCV infected patients who cannot clear the virus
- **broaden the T cell response;** both by making available new naïve T cells and by broadening the T cell repertoire
- may **foster the differentiation of protective memory** CD4 and CD8 T cells, which is typically impaired in chronic viral infections (PD-1)
- **anti-TGFβ** activity of IL-7 might significantly contribute to decreasing the excessive "T-reg" frequency.

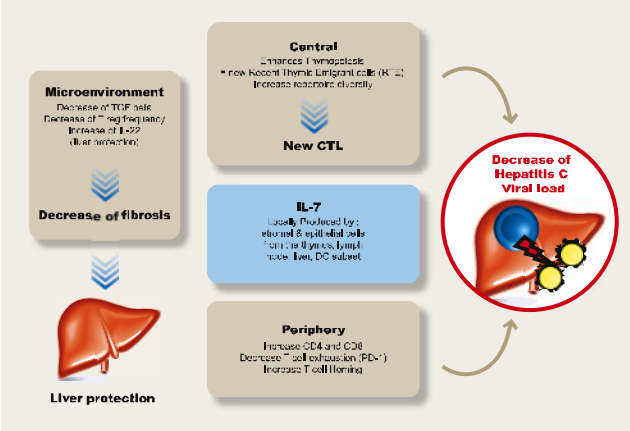
Microenvironment
Decrease of fibrosis
Liver protection
Central
New CTL
IL-7
Periphery
Decrease of Hepatitis C Viral load

Objectives

Evaluate the tolerance, immunological and antiviral effects of CYT107 in HCV-infected patients non-responders to Standard of Care (SoC)

➤ **Safety at W12 of biologically active doses of CYT107** added to pegylated interferon-alpha (PegIFN-α) and ribavirin (SoC)

➤ **Pharmacokinetics**

➤ **Immunological effects:**
- CD3, CD4, CD8 and subpopulations of lymphocytes (naive, memory, Treg)
- T cell Repertoire diversity
- T cell Homing

➤ **Antiviral effects**

Methods

Study design : open-label dose-escalating study multicentric phase I/II a

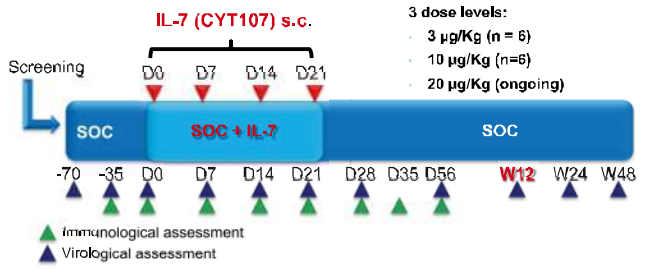
IL-7 (CYT107) s.c.
3 dose levels:
3 µg/Kg (n = 6)
10 µg/Kg (n=6)
20 µg/Kg (ongoing)
Screening
D0 D7 D14 D21
SOC
SOC + IL-7
SOC
-70 -35 D0 D7 D14 D21 D28 D35 D56 W12 W24 W48
Immunological assessment
Virological assessment

- **SoC** was initiated 6-10 weeks before initiation of CYT107, and continued as add-on during at least 12 weeks.
- **Assignment to a DL was doubled from 6 to 12 patients,**
 - if a reduction of HCV Viral Load ⩾ 2 log or HCV undetectable at W12 after CYT107 initiation (D0) was observed in at least 2 of 6 patients,
 - if more than 1 patient from the initial cohort at this DL experienced a DLT.

Patient Population and Baseline Demographics

Patient population

- **The treatment-experienced HCV (genotype 1 and 4) adult patients enrolled were prior non responders defined as:**
 - ➤ Absence of EVR , or
 - ➤ Absence of end of treatment response defined by detectable HCV RNA at the end of treatment (24 weeks or 48 weeks)
- **Key exclusion criteria included:**
 - ➤ Previous receipt of polymerase or protease inhibitor or any substance with immunological properties
 - ➤ Co-infection with HIV or HBV
 - ➤ Body mass index (BMI) > 30kg/m²
 - ➤ Auto-antibodies with a titer > 1/80
 - ➤ Alpha-fetoprotein > 200 ng/mL
 - ➤ ALT and/or AST > 5 x ULN (grade 2)
 - ➤ Any history of malignancy,
 - ➤ Auto-immune disease, transplantation.

Table 1 : Baseline patient demographics and disease characteristics

	3 µg/kg (n = 6)	10 µg/kg (n = 12)
Gender (F/M)	1/5	3/9
Race,		
• Caucasian	5	7
• North-African	-	3
• African	1	2
Age, years ; median (range)	47 (42 – 61)	50 (41 – 59)
BMI (Kg/m²), median (range)	24 (23 – 29)	26 (21 – 30)
HCV genotype (n)		
• Genotype 1	6	8
• Genotype 4	0	4
Total duration of previous SOC, wks; median	62 (16 – 108)	76 (14 – 286)
HCVRNA at baseline (Scr1), (log_{10} IU/ml), median (range)	6.29 (5.59 – 6.83)	6.17 (5.87 - 6.42)

Results

Presentation of the 3µg/kg and 10µg/Kg dose levels

Safety and tolerability

- One SUSAR (abdominal pain, grade 2) was considered as possibly related to CYT107 and/or to SoC. This SAE did not lead to the stop of the patient treatment.
- No other serious Adverse Events, DLT or clinically relevant abnormalities in biological parameters related to CYT107 treatment were reported.
- 78.6 % of AEs were of grade ≤1, primarily injection-site reactions.

Table 2 : Treatment Adverse Events (n patients, % of patients)

	3 µg/Kg/wk (N= 6)	10 µg/Kg/wk (N= 12)
• Local reactions at injection site (erythema, pruritus, induration, pain)	5 (83.3%)	11 (91.7%)
• Asthenia	0	3 (25%)
• Flu-like syndrome	0	1 (8.3%)
• Fever	0	2 (16.7%)
• Splenomegalia	0	1 (8.3%)
• Neutropenia	0	0
• Lymphocytopenia	0	1 (8.3%)

Figure 1 : T cell subsets evolution

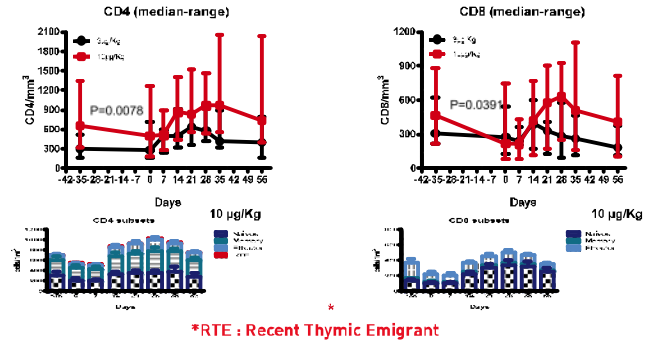
CD4 (median-range)
CD8 (median-range)
P=0.0078
P=0.0391
Days
10 µg/Kg
*RTE : Recent Thymic Emigrant

- CYT107 treatment led to a dose dependant increase of both CD4 and CD8 T cell
- All T cell subsets are increased.

Figure 2 : Repertoire diversity (10µ/Kg)

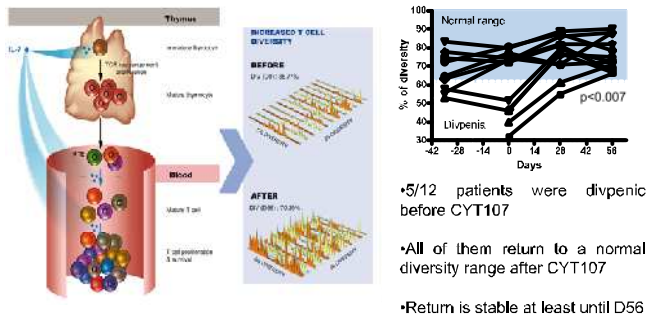
Normal range
p<0.007
Divpenis
Days
% of diversity

- 5/12 patients were divpenic before CYT107
- All of them return to a normal diversity range after CYT107
- Return is stable at least until D56

ImmunTraCkeR® test (ImmunID Technologies, Grenoble France). Briefly, 5.10⁶ PBMCs were thawed and resuspended in EasyID buffer containing proteinase K. Multiplex PCR was performed on the genomic DNA By using this technique, it is possible to detect all the possible TRBVJ combinatorial rearrangements.

Figure 3 : T cell homing (α4β7) (10µ/Kg)

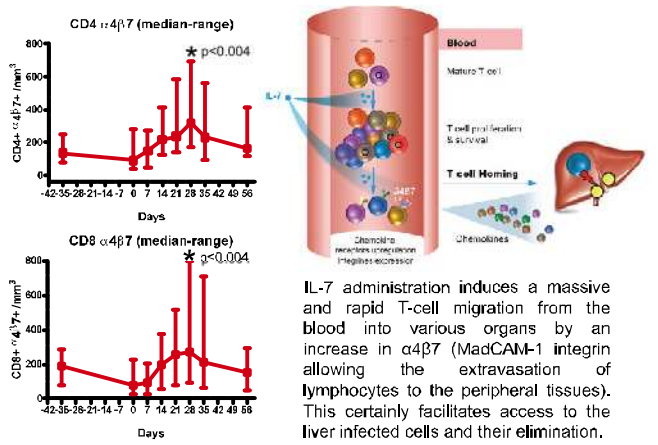
CD4 α4β7 (median-range)
* p<0.004
CD8 α4β7 (median-range)
* p<0.004
Days
Blood
T cell Homing
Chemokines

IL-7 administration induces a massive and rapid T-cell migration from the blood into various organs by an increase in α4β7 (MadCAM-1 integrin allowing the extravasation of lymphocytes to the peripheral tissues). This certainly facilitates access to the liver infected cells and their elimination.

Figure 4 : Viral load

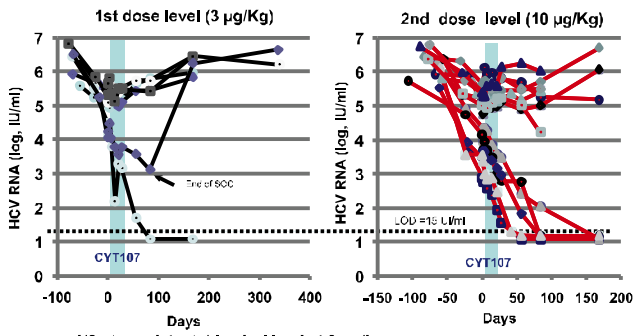
1st dose level (3 µg/Kg)
2nd dose level (10 µg/Kg)
HCV RNA (log. IU/ml)
CYT107
End of SOC
LOD =15 UI/ml
Days

- 1/6 pts undetectable viral load at 3 µg/kg
- 5/12 pts undetectable viral load at 10 µg/Kg
 - Genotype 1: n=4
 - Genotype 4: n=1

Figure 5 : Comparison of Viral Load Changes (log/day)-CYT107 vs. SoC

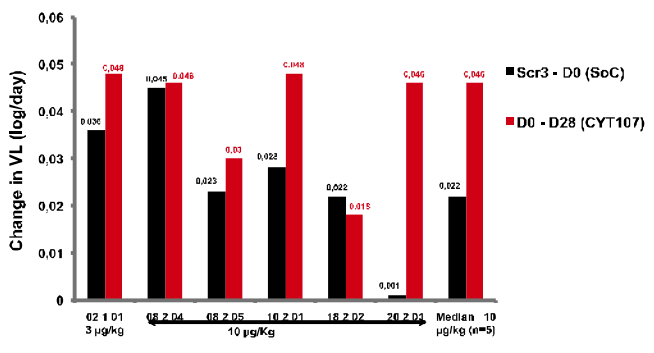
Change in VL (log/day)
Scr3 - D0 (SoC)
D0 - D28 (CYT107)
3 µg/kg
10 µg/Kg
Median 10 µg/kg (n=5)

Conclusion: the slopes of viral decrease were larger after CYT107 administrations (D0-D28) than during the SoC treatment (Scr3 -D0)

Conclusions

In chronic HCV- patients, non-responders to pegIFN + RBV,

- CYT107 was generally well-tolerated when co-administered for 4 weeks to pegIFN/RBV
- CYT107 expanded both CD4 and CD8 T cells, an effect known to provide an efficient and stable immune response,
- CYT107 contributed to an increase of T cell homing in lymphoid organs
- CYT107 induced a normalization of the diversity of the TCR repertoire in most patients
- At 10µg/kg/wk in combination with pegIFN and RBV, these immune effects were associated with a HCVRNA drop to undetectable levels in a significant number of patients.

These results suggest the need for future studies combining DAAs and CYT107 in patients with CHC.

References

1. Klenerman P et al. Gut. 2011 ;
2. Pellegrini M, et al. Cell. 2011 ;
3. Sportès C et al. Ann N Y Acad Sci. 2009 .
4. Levy Y, et al. J Clin Invest. 2009 ;
5. Sportès C etal. J Exp Med. 2008 ;
6. Nanjappa SG et al. Blood 2001;117:5123-32.

Acknowledgements

We thank all the study participants, the study investigative teams and Cytheris collaborators.

Presented at the 62nd Annual Meeting of the American Association for the Study of Liver Diseases, November 4-8, San Francisco